POSTER #2127

RELATED OPERA POSTERS: 1512, 1517, 1541

Predictors of Virologic Success in an Older ART Naïve HIV Population

Michael Wohlfeiler[1], Cassidy Henegar[2], Kathy Schulman[3], Jennifer Fusco[2], Susan Zelt[4], Ronald D'Amico[4], and George Melikian[1]

[1]AIDS Healthcare Foundation, Los Angeles, CA; [2]Epividian, Inc., Durham, NC; [3]Outcomes Research Solutions, Waltham, MA; [4]Viiv Healthcare Inc., Research Triangle Park, NC

OPERA® The Longitudinal Cohort

Contact Information:
4819 Emperor Blvd., Suite 400 Durham, NC 27703
P: 919-825-3457 F: 919-313-4505
Email: jennifer.fusco@epividian.com

BACKGROUND

The CDC reports that nearly a quarter of all individuals living with HIV are 50 years or older and a fifth of all new infections are in this age group.[1,2] Older HIV+ patients are more likely to be balancing HIV treatment with the challenges of aging (senescence, comorbidities, polypharmacy) than younger patients. While their risk factors for infection are similar, their predictors of treatment success may be quite different.

OBJECTIVE:

To identify and compare predictors of virologic suppression in treatment-naïve people 26-49 years old versus people 50 or more years old with HIV.

METHODS

- The study population was selected from the Observational Pharmaco-Epidemiology Research and Analysis (OPERA) cohort, which includes prospectively-captured, routine clinical data from patients at 79 outpatient clinics in 15 states in the United States.

Figure 1. Geographic Distribution of OPERA Cohort

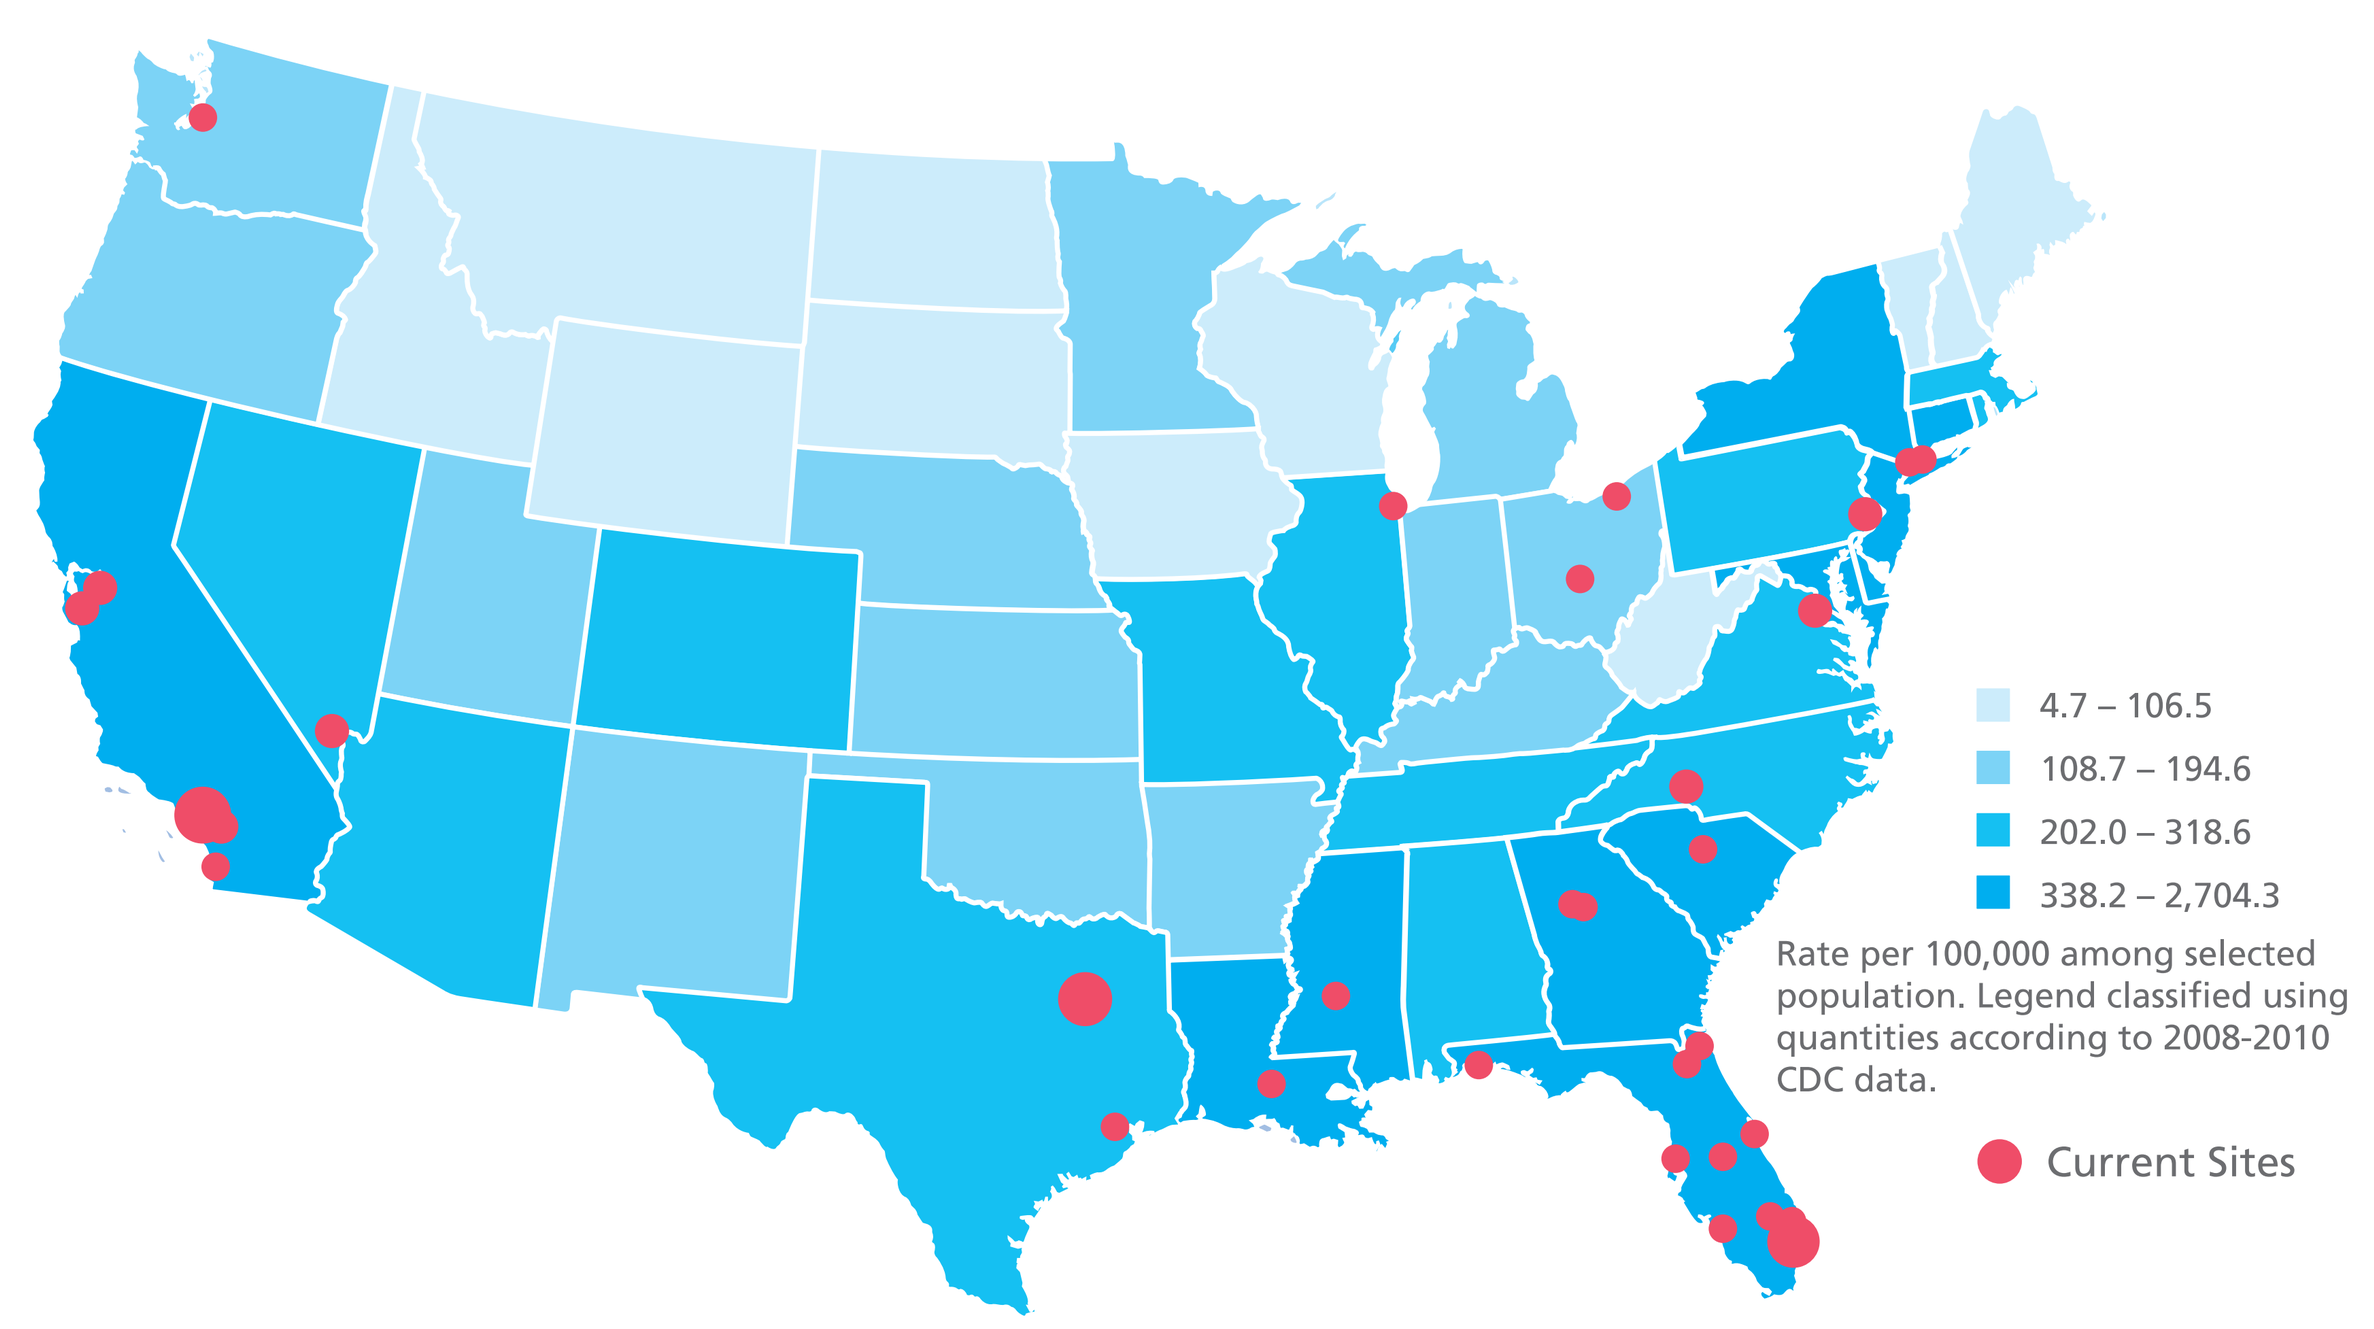
4.7 – 106.5
108.7 – 194.6
202.0 – 318.6
338.2 – 2,704.3
Rate per 100,000 among selected population. Legend classified using quantities according to 2008-2010 CDC data.
Current Sites

- Patients eligible for analysis included treatment-naïve HIV+ individuals starting their first ART regimen between January 1, 2007 and March 31, 2015, and who were at least 26 years old at the time of ART initiation. Adolescent and young adults were analyzed seperately (see IDWeek poster #1541).
- Patients were followed from the start date of their first ART regimen until any regimen change, death, loss to follow-up, or study end (March 31, 2016).
- Treatment outcomes:
- Virologic suppression was defined as a single HIV-1 RNA viral load (VL) of <50 copies/mL.
- Virologic rebound could occur only in patients who first achieved suppression. Rebound was defined as either of the following:
 - 1 VL >200 copies/mL followed by regimen discontinuation OR
 - 2 VL >200 copies/mL
- Cox proportional hazards regression was used to simultaneously assess the association between several patient characteristics (measured at baseline) and virologic suppression during an initial ART regimen. Separate models were created and compared for 26-49 year olds and 50+ year olds.

RESULTS

Table 1: Selected Baseline* Clinical Characteristics

	Patients 26-49 years N= 6139	Patients 50+ years N= 1174	p-value
Male	5264 (85.8%)	891 (75.9%)	**<0.0001**
African American	2125 (34.6%)	500 (42.6%)	**<0.0001**
Hispanic	1624 (26.5%)	169 (14.4%)	**<0.0001**
MSM+	3545 (57.7%)	452 (38.5%)	**<0.0001**
Treated in Southeast US	2793 (45.5%)	671 (57.2%)	**<0.0001**
Baseline Viral Load			
Low (<10,000 copies/mL)	1015 (16.5%)	185 (15.8%)	**<0.36**
Moderate (≥10,000 to <100,000 copies/mL)	3032 (49.4%)	564 (48.0%)	
High (≥100,000 copies/mL)	2092 (34.1%)	425 (36.2%)	
Baseline CD4			
Early presenter (>500 cells/μL)	1236 (20.1%)	202 (17.2%)	**<0.0001**
Moderate presenter (>350 to ≤500 cells/μL)	1415 (23.0%)	221 (18.8%)	
Late presenter (>200 to ≤350 cells/μL)	1717 (28.0%)	313 (26.7%)	
Later presenter (>50 to ≤200 cells/μL)	1153 (18.8%)	290 (24.7%)	
Latest presenter (≤50 cells/μL)	618 (10.1%)	148 (12.6%)	
AIDS at baseline	474 (7.7%)	117 (10.0%)	**0.001**
Year of ART initiation			
2007-2011	2668 (43.5%)	473 (40.3%)	**<0.0001**
2012-2015	3471 (56.5%)	701 (59.7%)	
Hepatitis C ever at baseline	336 (5.5%)	188 (16.0%)	**<0.0001**
Syphilis ever at baseline	1640 (26.7%)	239 (20.4%)	**<0.0001**
Non-HIV pill burden			
0-4 pills per day	5443 (88.7%)	901 (76.7%)	**<0.0001**
5-9 pills per day	556 (9.1%)	215 (18.3%)	
10+ pills per day	140 (2.3%)	58 (4.9%)	
Number of comorbid conditions			**<0.0001**
0 conditions	3589 (58.5%)	410 (34.9%)	
1 condition	1778 (29.0%)	352 (30.0%)	
2+ conditions	772 (12.6%)	412 (35.1%)	

* Baseline: Initiation of HIV ART; + MSM: men who have sex with men

Figure 2. Pill Burden And Core Agent Drug Class By Age Group

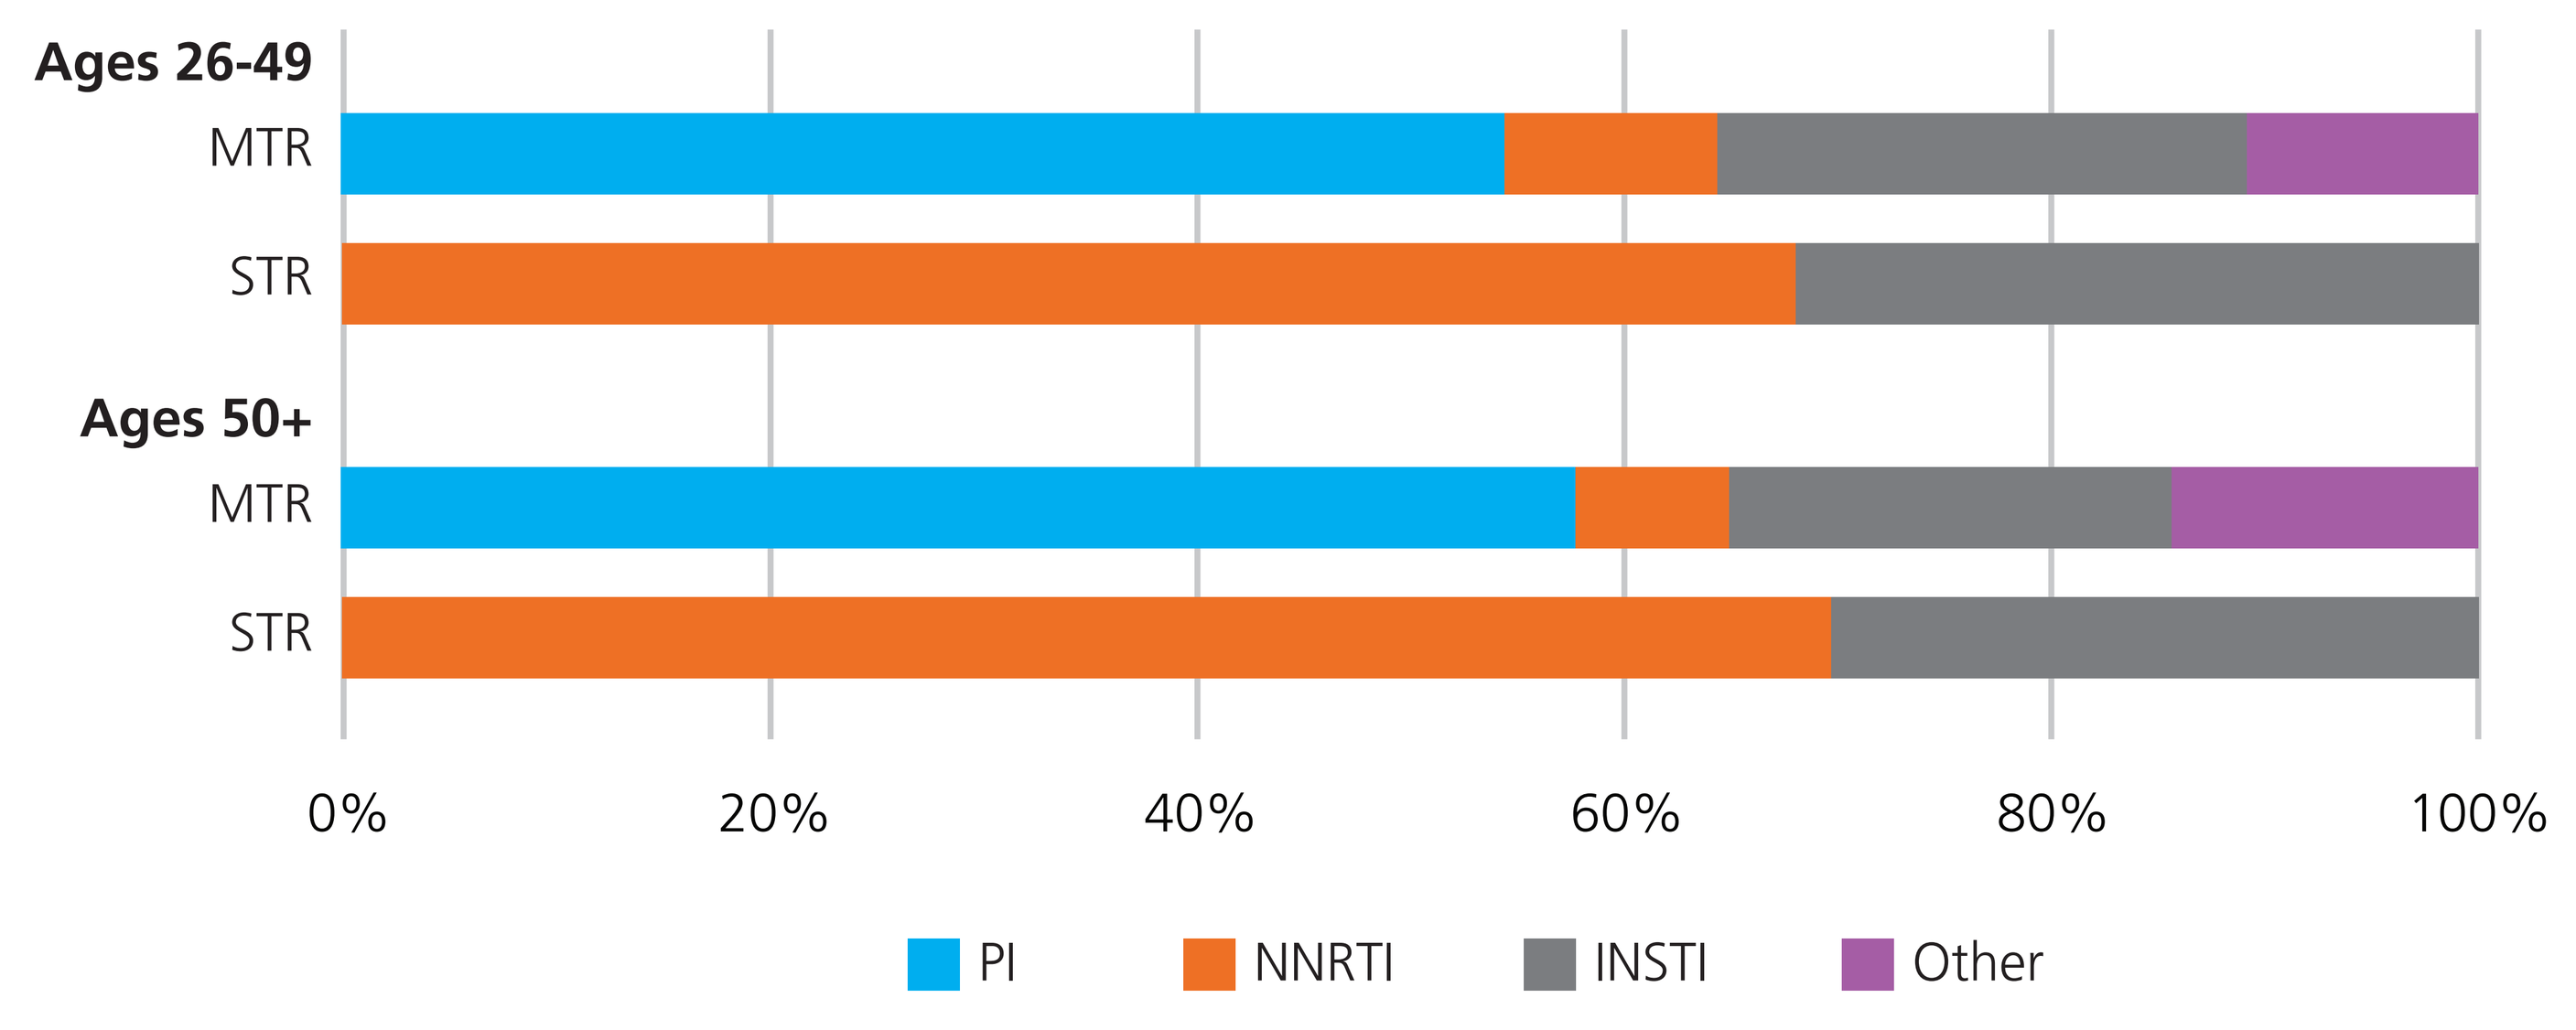
Ages 26-49
MTR
STR
Ages 50+
MTR
STR
0%
20%
40%
60%
80%
100%
PI
NNRTI
INSTI
Other

- At the time of ART initiation, patients 50 or older had lower CD4 counts, were more likely to have a history of AIDS-defining event or Hepatitis C infection, had a higher number of chronic comorbidities, and took more non-ART medications each day.
- Initiating with an STR was more common among 26-49 year olds (59% vs. 48%; p<0.0001).

Table 2: Treatment Outcomes Of Initial ART Regimen By Age Groups

	Patients 26-49 years N= 6139	Patients 50+ years N= 1174	p-value
Virologic Suppression			
Achieved virologic suppression (<50 copies/mL) during initial ART regimen	3897 (63.5%)	738 (62.9%)	0.69
Had at least one VL after baseline	5288 (86.1%)	1022 (87.1%)	0.40
Patients with VLs: Achieved virologic suppression	3897 (73.7%)	738 (72.2%)	0.33
Time to first VL < 50 copies/mL [median (IQR) months]	4.1 (2.3, 6.5)	4.0 (2.4, 6.0)	0.17
Virologic Rebound			
2 X (VL >200 copies/mL) after suppression	331 (5.4%)	62 (5.3%)	0.88
1 X (VL >200 copies/mL) after suppression plus discontinuation of initial ART regimen	248 (4.0%)	45 (3.8%)	0.74
Time to virologic rebound [median (IQR) months]	17.1 (10.9, 27.1)	14.5 (9.2, 19.5)	**0.0006**
Discontinuation			
Discontinued initial ART regimen during follow-up period	4107 (66.9%)	827 (70.4%)	**<0.0001**
Time to discontinuation [median (IQR) months]	12.0 (5.1, 24.3)	12.0 (4.3, 20.7)	**0.0006**

- Similar proportions of younger and older adults achieved virologic suppression on their initial ART regimen. Among patients who achieved suppression, about 9% of patients in each age group (26-49 and 50+) went on to experience an event of virologic rebound. Time to rebound was shorter for patients 50 or older.
- More patients 50+ discontinued their initial ART regimen during the observational period.

Figure 3: Association Of Patient Demographic And Clinical Characteristics With Virologic Suppression Among HIV-Positive, Treatment-Naïve Patients Initiating ART

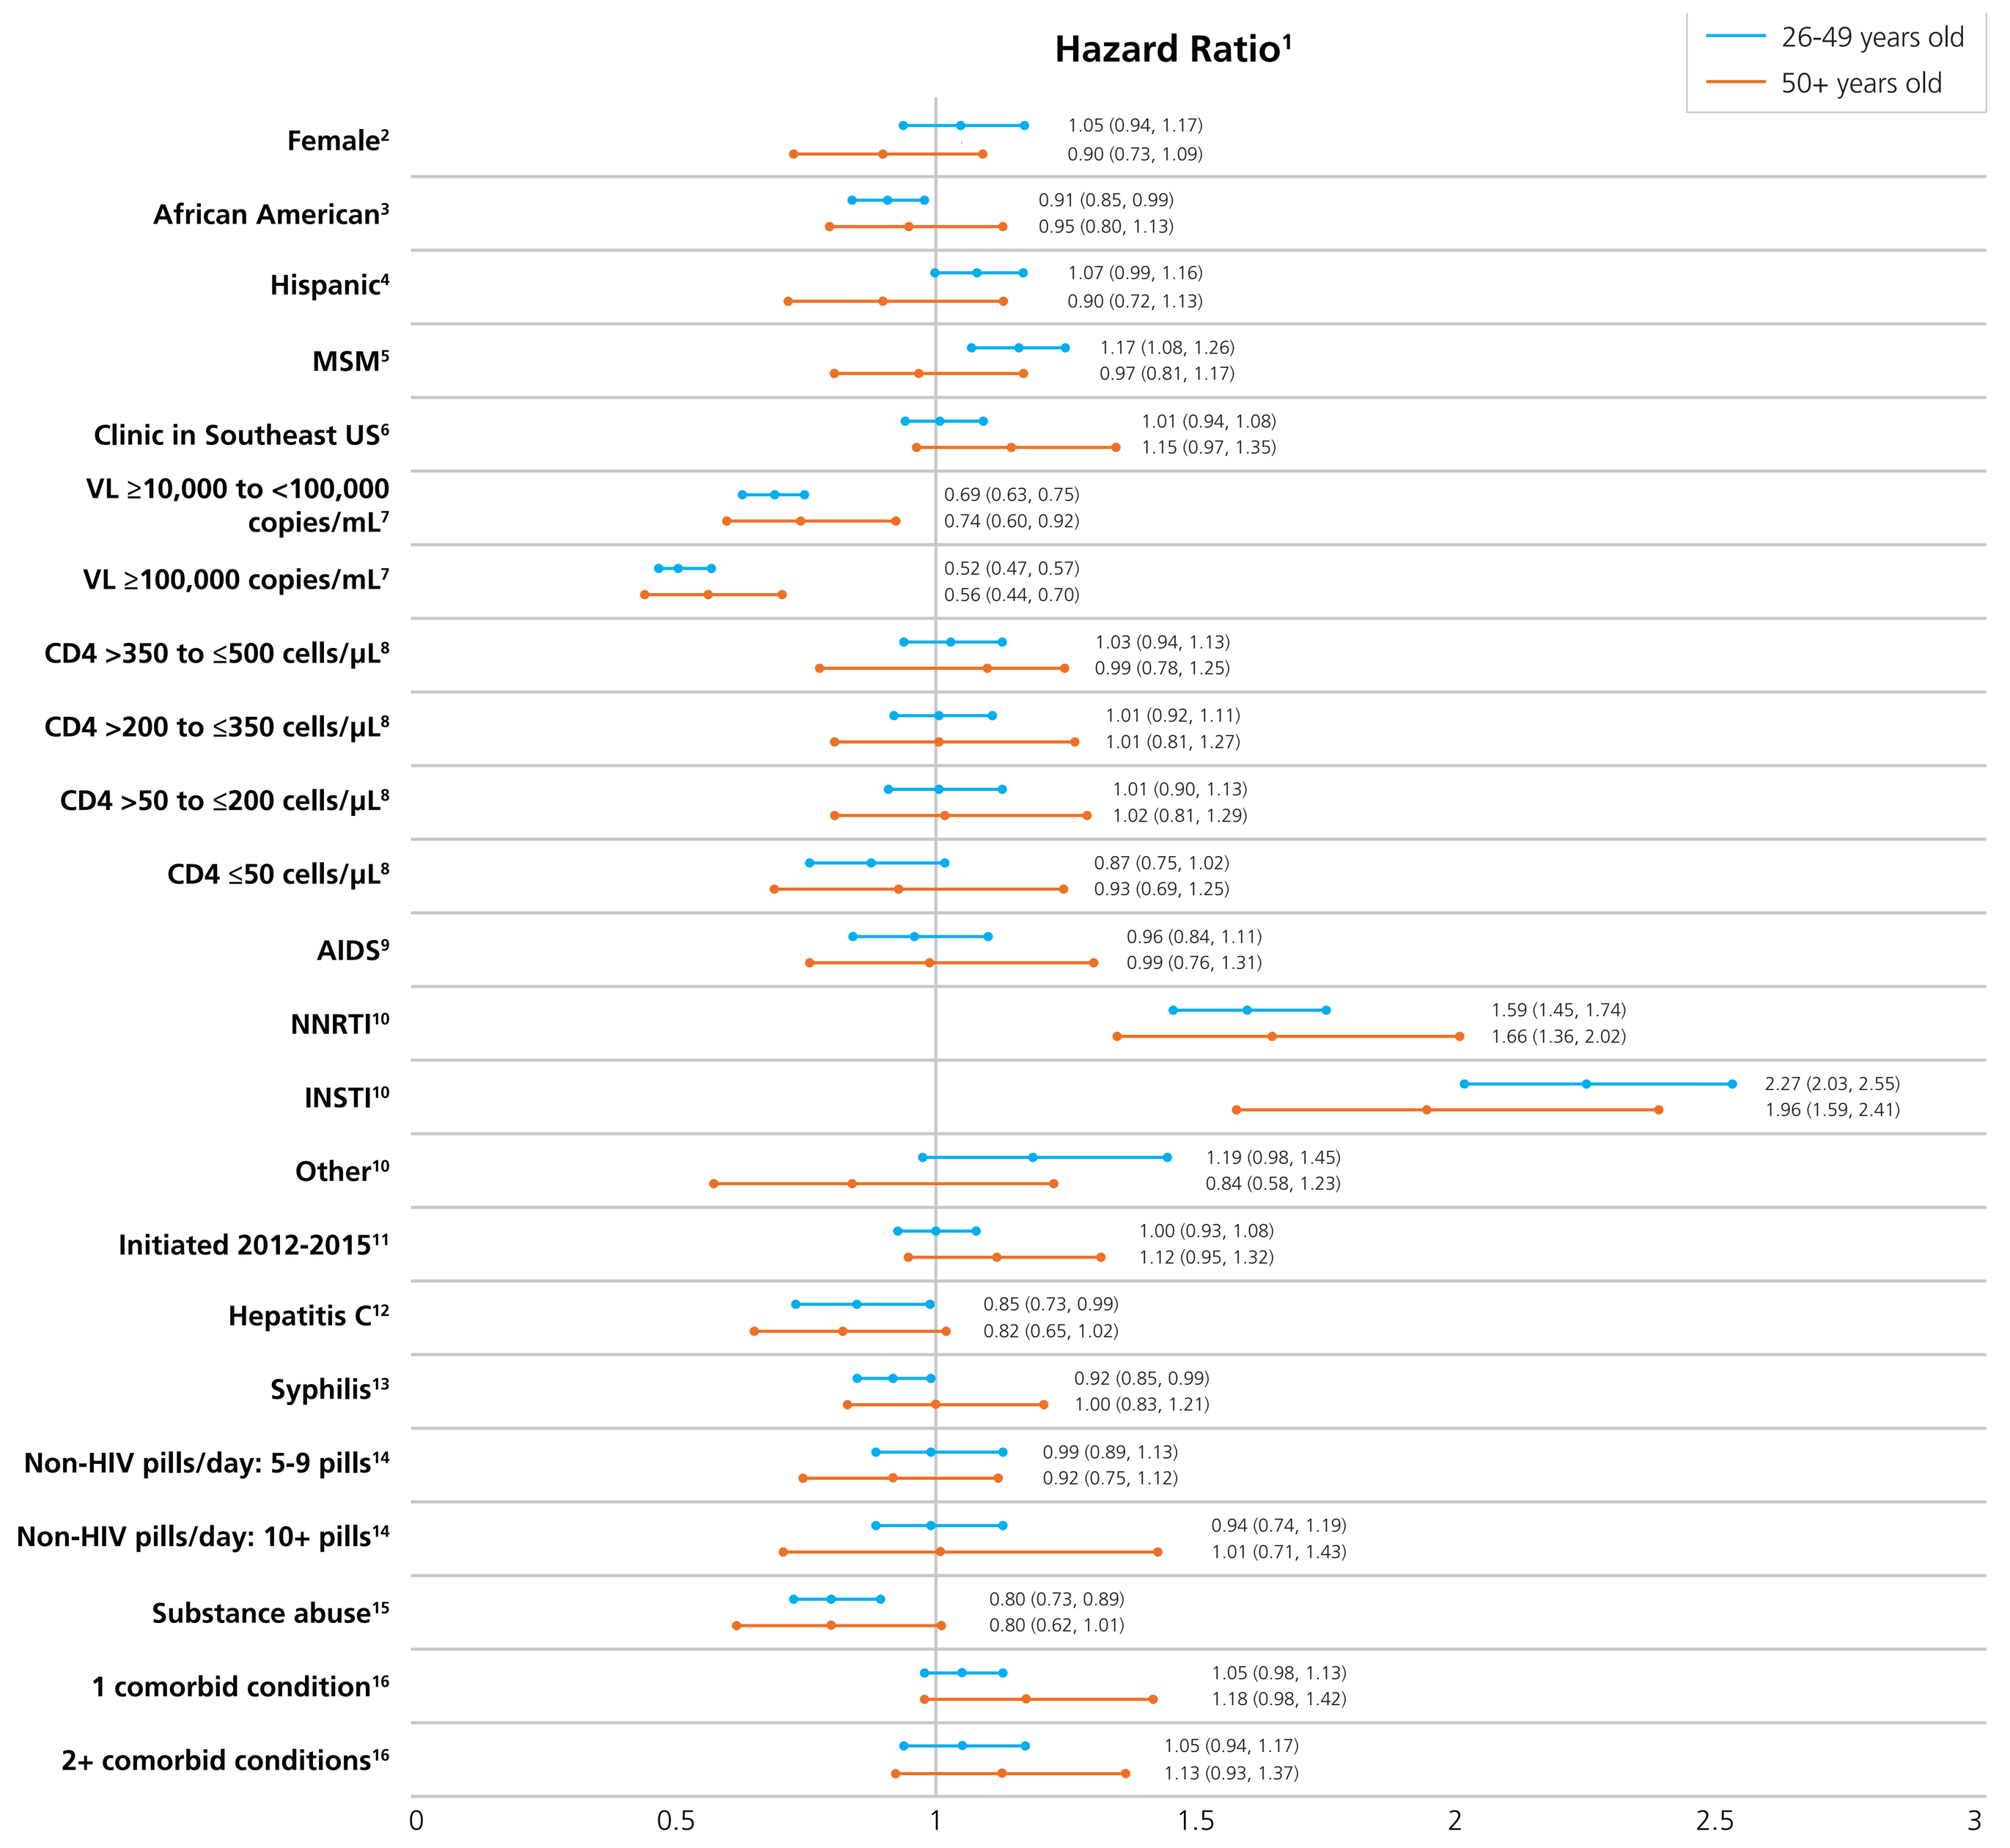
Hazard Ratio¹
26-49 years old
50+ years old
Female² 1.05 (0.94, 1.17) 0.90 (0.73, 1.09)
African American³ 0.91 (0.85, 0.99) 0.95 (0.80, 1.13)
Hispanic⁴ 1.07 (0.99, 1.16) 0.90 (0.72, 1.13)
MSM⁵ 1.17 (1.08, 1.26) 0.97 (0.81, 1.17)
Clinic in Southeast US⁶ 1.01 (0.94, 1.08) 1.15 (0.97, 1.35)
VL ≥10,000 to <100,000 copies/mL⁷ 0.69 (0.63, 0.75) 0.74 (0.60, 0.92)
VL ≥100,000 copies/mL⁷ 0.52 (0.47, 0.57) 0.56 (0.44, 0.70)
CD4 >350 to ≤500 cells/μL⁸ 1.03 (0.94, 1.13) 0.99 (0.78, 1.25)
CD4 >200 to ≤350 cells/μL⁸ 1.01 (0.92, 1.11) 1.01 (0.81, 1.27)
CD4 >50 to ≤200 cells/μL⁸ 1.01 (0.90, 1.13) 1.02 (0.81, 1.29)
CD4 ≤50 cells/μL⁸ 0.87 (0.75, 1.02) 0.93 (0.69, 1.25)
AIDS⁹ 0.96 (0.84, 1.11) 0.99 (0.76, 1.31)
NNRTI¹⁰ 1.59 (1.45, 1.74) 1.66 (1.36, 2.02)
INSTI¹⁰ 2.27 (2.03, 2.55) 1.96 (1.59, 2.41)
Other¹⁰ 1.19 (0.98, 1.45) 0.84 (0.58, 1.23)
Initiated 2012-2015¹¹ 1.00 (0.93, 1.08) 1.12 (0.95, 1.32)
Hepatitis C¹² 0.85 (0.73, 0.99) 0.82 (0.65, 1.02)
Syphilis¹³ 0.92 (0.85, 0.99) 1.00 (0.83, 1.21)
Non-HIV pills/day: 5-9 pills¹⁴ 0.99 (0.89, 1.13) 0.92 (0.75, 1.12)
Non-HIV pills/day: 10+ pills¹⁴ 0.94 (0.74, 1.19) 1.01 (0.71, 1.43)
Substance abuse¹⁵ 0.80 (0.73, 0.89) 0.80 (0.62, 1.01)
1 comorbid condition¹⁶ 1.05 (0.98, 1.13) 1.18 (0.98, 1.42)
2+ comorbid conditions¹⁶ 1.05 (0.94, 1.17) 1.13 (0.93, 1.37)
0
0.5
1
1.5
2
2.5
3

1. Hazard ratio >1.0 indicates increased probability of achieving virologic suppression

Referent category: 2. Male, 3. Not African American, 4. Not Hispanic, 5. Not a man who has sex with men (MSM), 6. Clinic in any other geographic region (Northeast, Mid-Atlantic, Midwest, Southwest, West), 7. VL <10,000 copies/mL, 8. CD4 >500 cells/μL, 9. No diagnosis of AIDS at baseline, 10. Initiated on a protease inhibitor-containing regimen, 11. Initiated ART 2007-2011, 12. No history of HCV at baseline, 13. No history of syphilis at baseline, 14. 0-4 non-ART pills/day, 15. no history of substance abuse at baseline, 16. history of 0 comorbid conditions at baseline

- Regardless of age, baseline VL and drug class of the anchor agent in the initial ART regimen were the strongest predictors of virologic suppression.
 - Compared to PIs, patients taking INSTI-based regimens were the most likely to achieve suppression, followed by patients taking NNRTI-based regimens.
- Baseline CD4 count was not associated with viral suppression for either age group.
- Having a history of hepatitis C, syphilis, or substance abuse was associated with lower probability of suppression for adults 26-49, but not for older adults (50+).
- Even though they were taking more non-ART pills per day, older adults (50+) were prescribed more multiple tablet regimens containing 3 or more pills per day.

DISCUSSION

- Older patients (50+ years) shared several predictors of virologic success with younger adults (26-49 years), including baseline VL and type of ART initiated, despite initiating with significantly different baseline demographics and clinical characteristics.
- While patients 50 and older were dealing with more comorbidities and taking more pills per day, these complexities did not appear to impact the likelihood of older patients to suppress HIV viral load. Further analysis of specific conditions and medication classes, rather than simple counts of comorbidities and pills, may provide more insight into how dealing with other medical conditions and medications while starting HIV therapy impacts treatment outcomes.
- While initial ability to suppress viral load after starting ART did not vary much between age groups, older patients appeared to be less likely to maintain suppression and stay on their initial ART regimens. These areas require additional research.

KEY FINDING:

In a large US HIV database, early diagnosis, baseline viral load, and access to INSTIs or NNRTIs at ART initiation were the most important factors predicting virologic success in older HIV+ individuals.

ACKNOWLEDGMENTS

This research would not be possible without the generosity of the OPERA HIV caregivers and their patients sharing their experiences. Additionally, we are grateful for the following individuals: Gregory Fusco MD MPH (Advisory Board coordination), Robin Beckerman (SAS programming), Ted Ising (Database Arch & Mgmt), Bernie Stooks (Database Mgmt), Judy Johnson (Med Terminology Classification), Rodney Mood (Site Support & Data Analyst), Elizabeth Yancy (Data Analyst).

REFERENCES

1. CDC: HIV Among People Aged 50 and Over. Atlanta, Georgia: Centers for Disease Control and Prevention (2016); Accessed September 8, 2016.
2. CDC: Diagnoses of HIV infection in the United States and dependent areas; 2013. HIV Surveillance Report 2015: 25. Accessed September 8, 2016.

SPONSORSHIP

This research was funded by ViiV Healthcare

ViiV Healthcare

Epividian®